356

Obeticholic Acid Improves Histological, Biochemical and Gene Expression Profiles in Gubra AMLN Mice with Diet-Induced and Biopsy-Confirmed NASH

Jonathan Roth[1], Michael Feigh[2], Sanne Veidal[2], Kristoffer Rigbolt[2], Mark Young[1]

1. Intercept Pharmaceuticals, Inc., San Diego, CA, United States; 2. Gubra, Hørsholm, Denmark

Intercept

STUDY DESIGN AND METHODS

Tissue Distribution Study

- Chow-fed C57Bl/6 mice (n=8/group) were treated with vehicle (0.5% CMC, PO, QD) or OCA (1, 3, 10 or 30 mg/kg) for 14 days
- Liver and ileum were analyzed for levels of OCA and its tauro- and glyco-conjugates by LC/MS/MS

NASH Study Outline

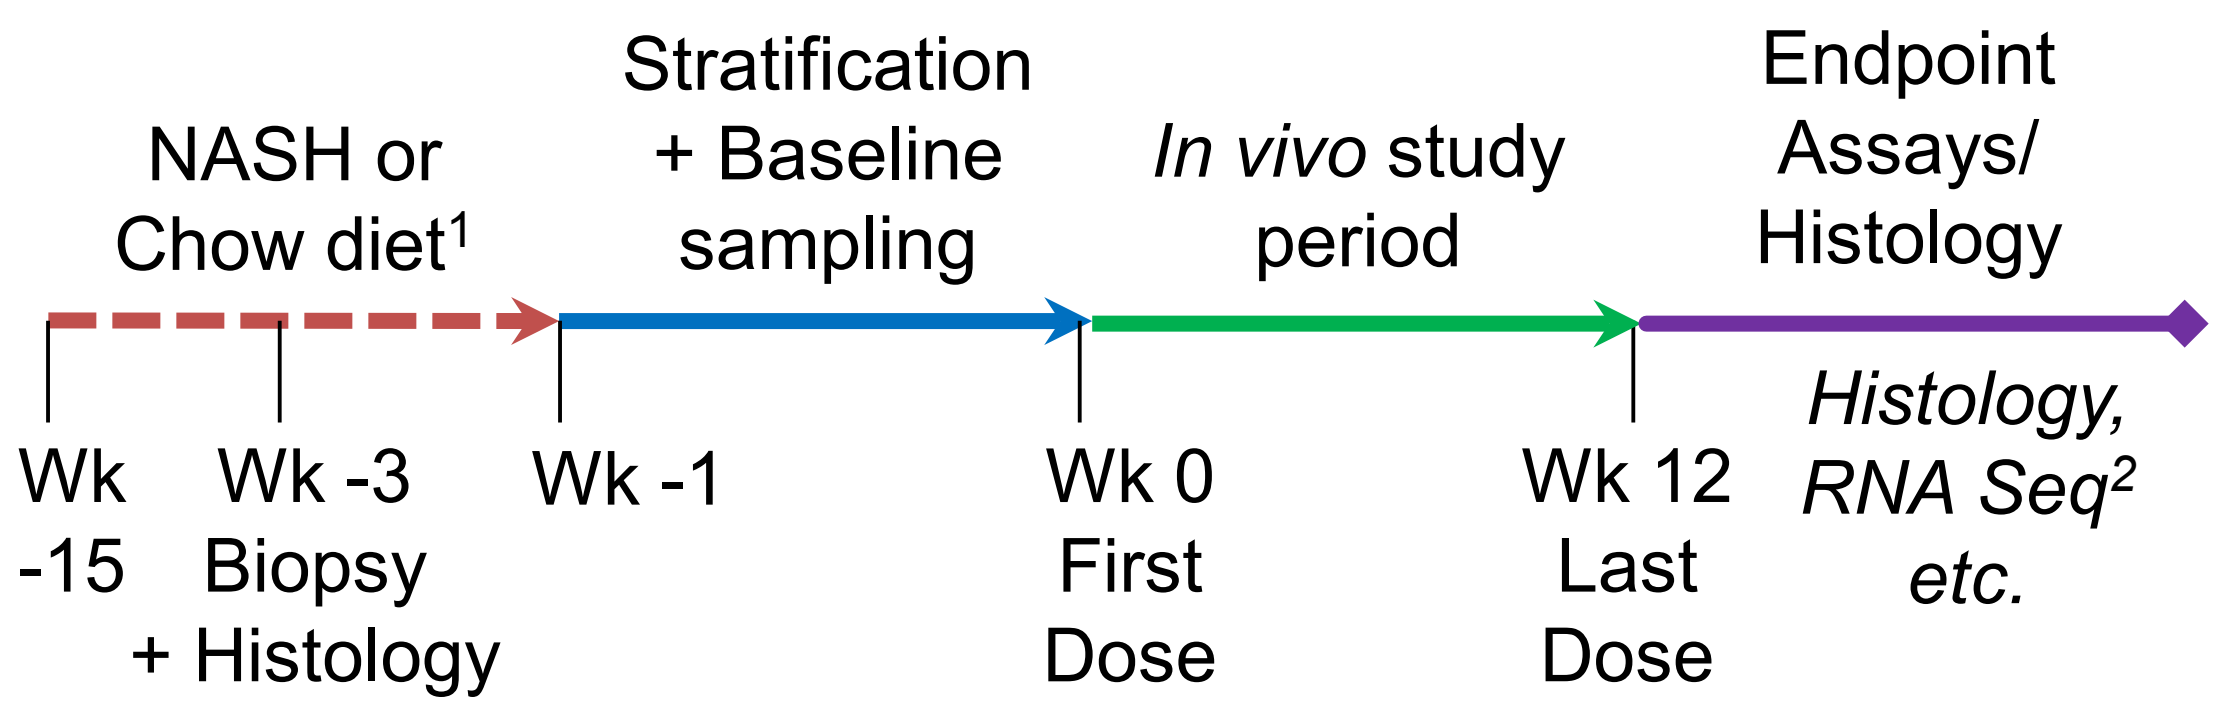

NASH or Chow diet[1]
Stratification + Baseline sampling
In vivo study period
Endpoint Assays/ Histology
Wk -15
Wk -3 Biopsy + Histology
Wk -1
Wk 0 First Dose
Wk 12 Last Dose
Histology, RNA Seq[2] *etc.*

[1]Mice remained on their respective diet for duration of study.
[2]mRNA data from mice treated for 2 weeks (Vehicle or OCA 10 mg/kg).

Liver and Ileum Gene Expression (NASH Study)

- Liver tissue from left lateral lobe, snap frozen, homogenized and RNA was extracted
- RNA Seq libraries were prepared with the KAPA poly-A kit, sequenced on the NextSeq 500 (Illumina)
- Reads were aligned to GRCm38 Ensembl Mus musculus genome using STAR v.2.4.0 and feature counts obtained using Htseq v.0.6.1

RESULTS

OCA Distribution in the Liver and Ileum

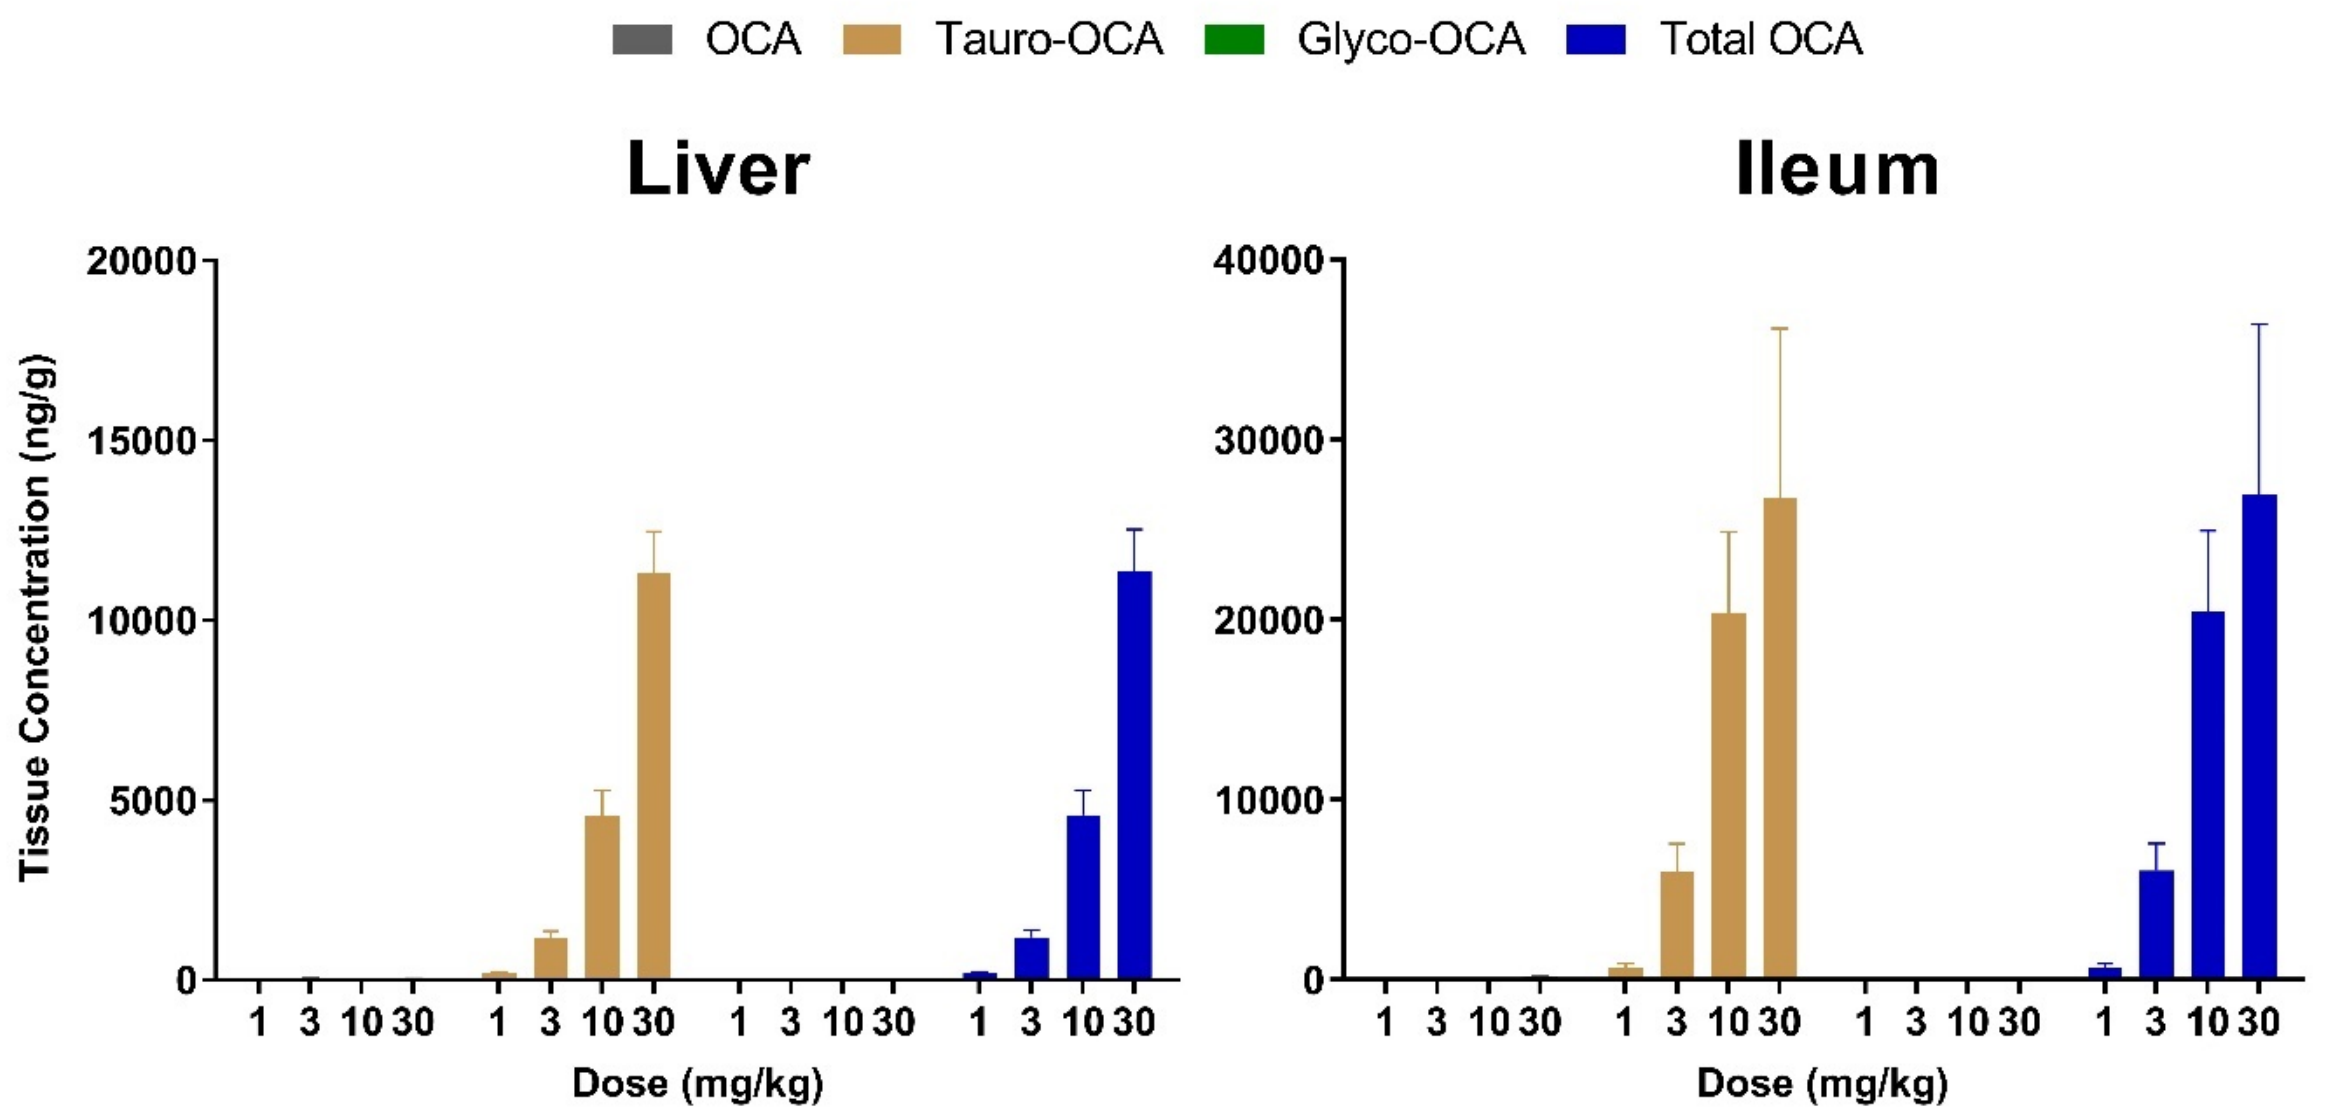

OCA
Tauro-OCA
Glyco-OCA
Total OCA
Liver
Ileum
Tissue Concentration (ng/g)
Dose (mg/kg)

Data are Mean ± SEM.

OCA Decreases Liver Weight†

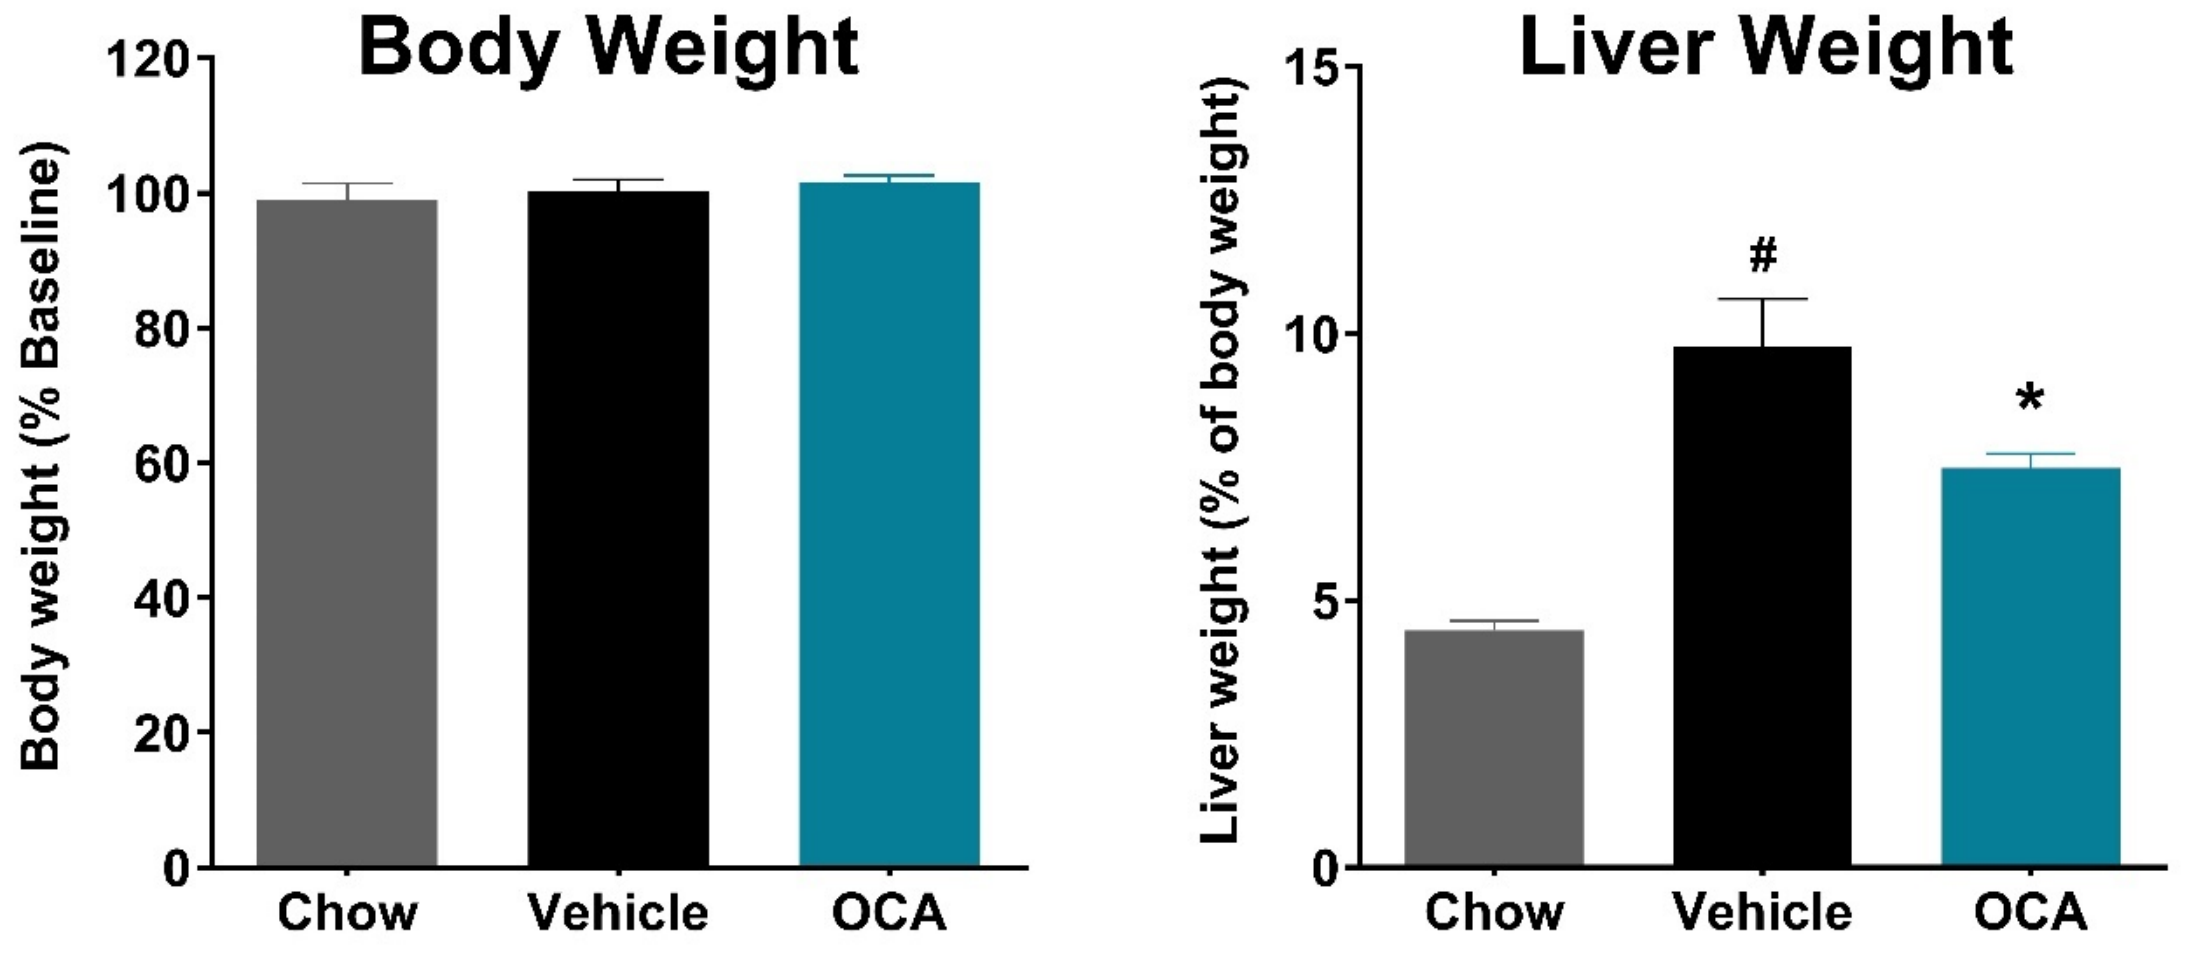

Body Weight
Body weight (% Baseline)
Liver Weight
Liver weight (% of body weight)
Chow
Vehicle
OCA

†Data are Mean ± SEM; #p<0.05 Vehicle vs Chow; *p<0.05 OCA vs Vehicle, ANOVA with Dunnett's post-hoc.

RESULTS

Representative Liver Photomicrographs

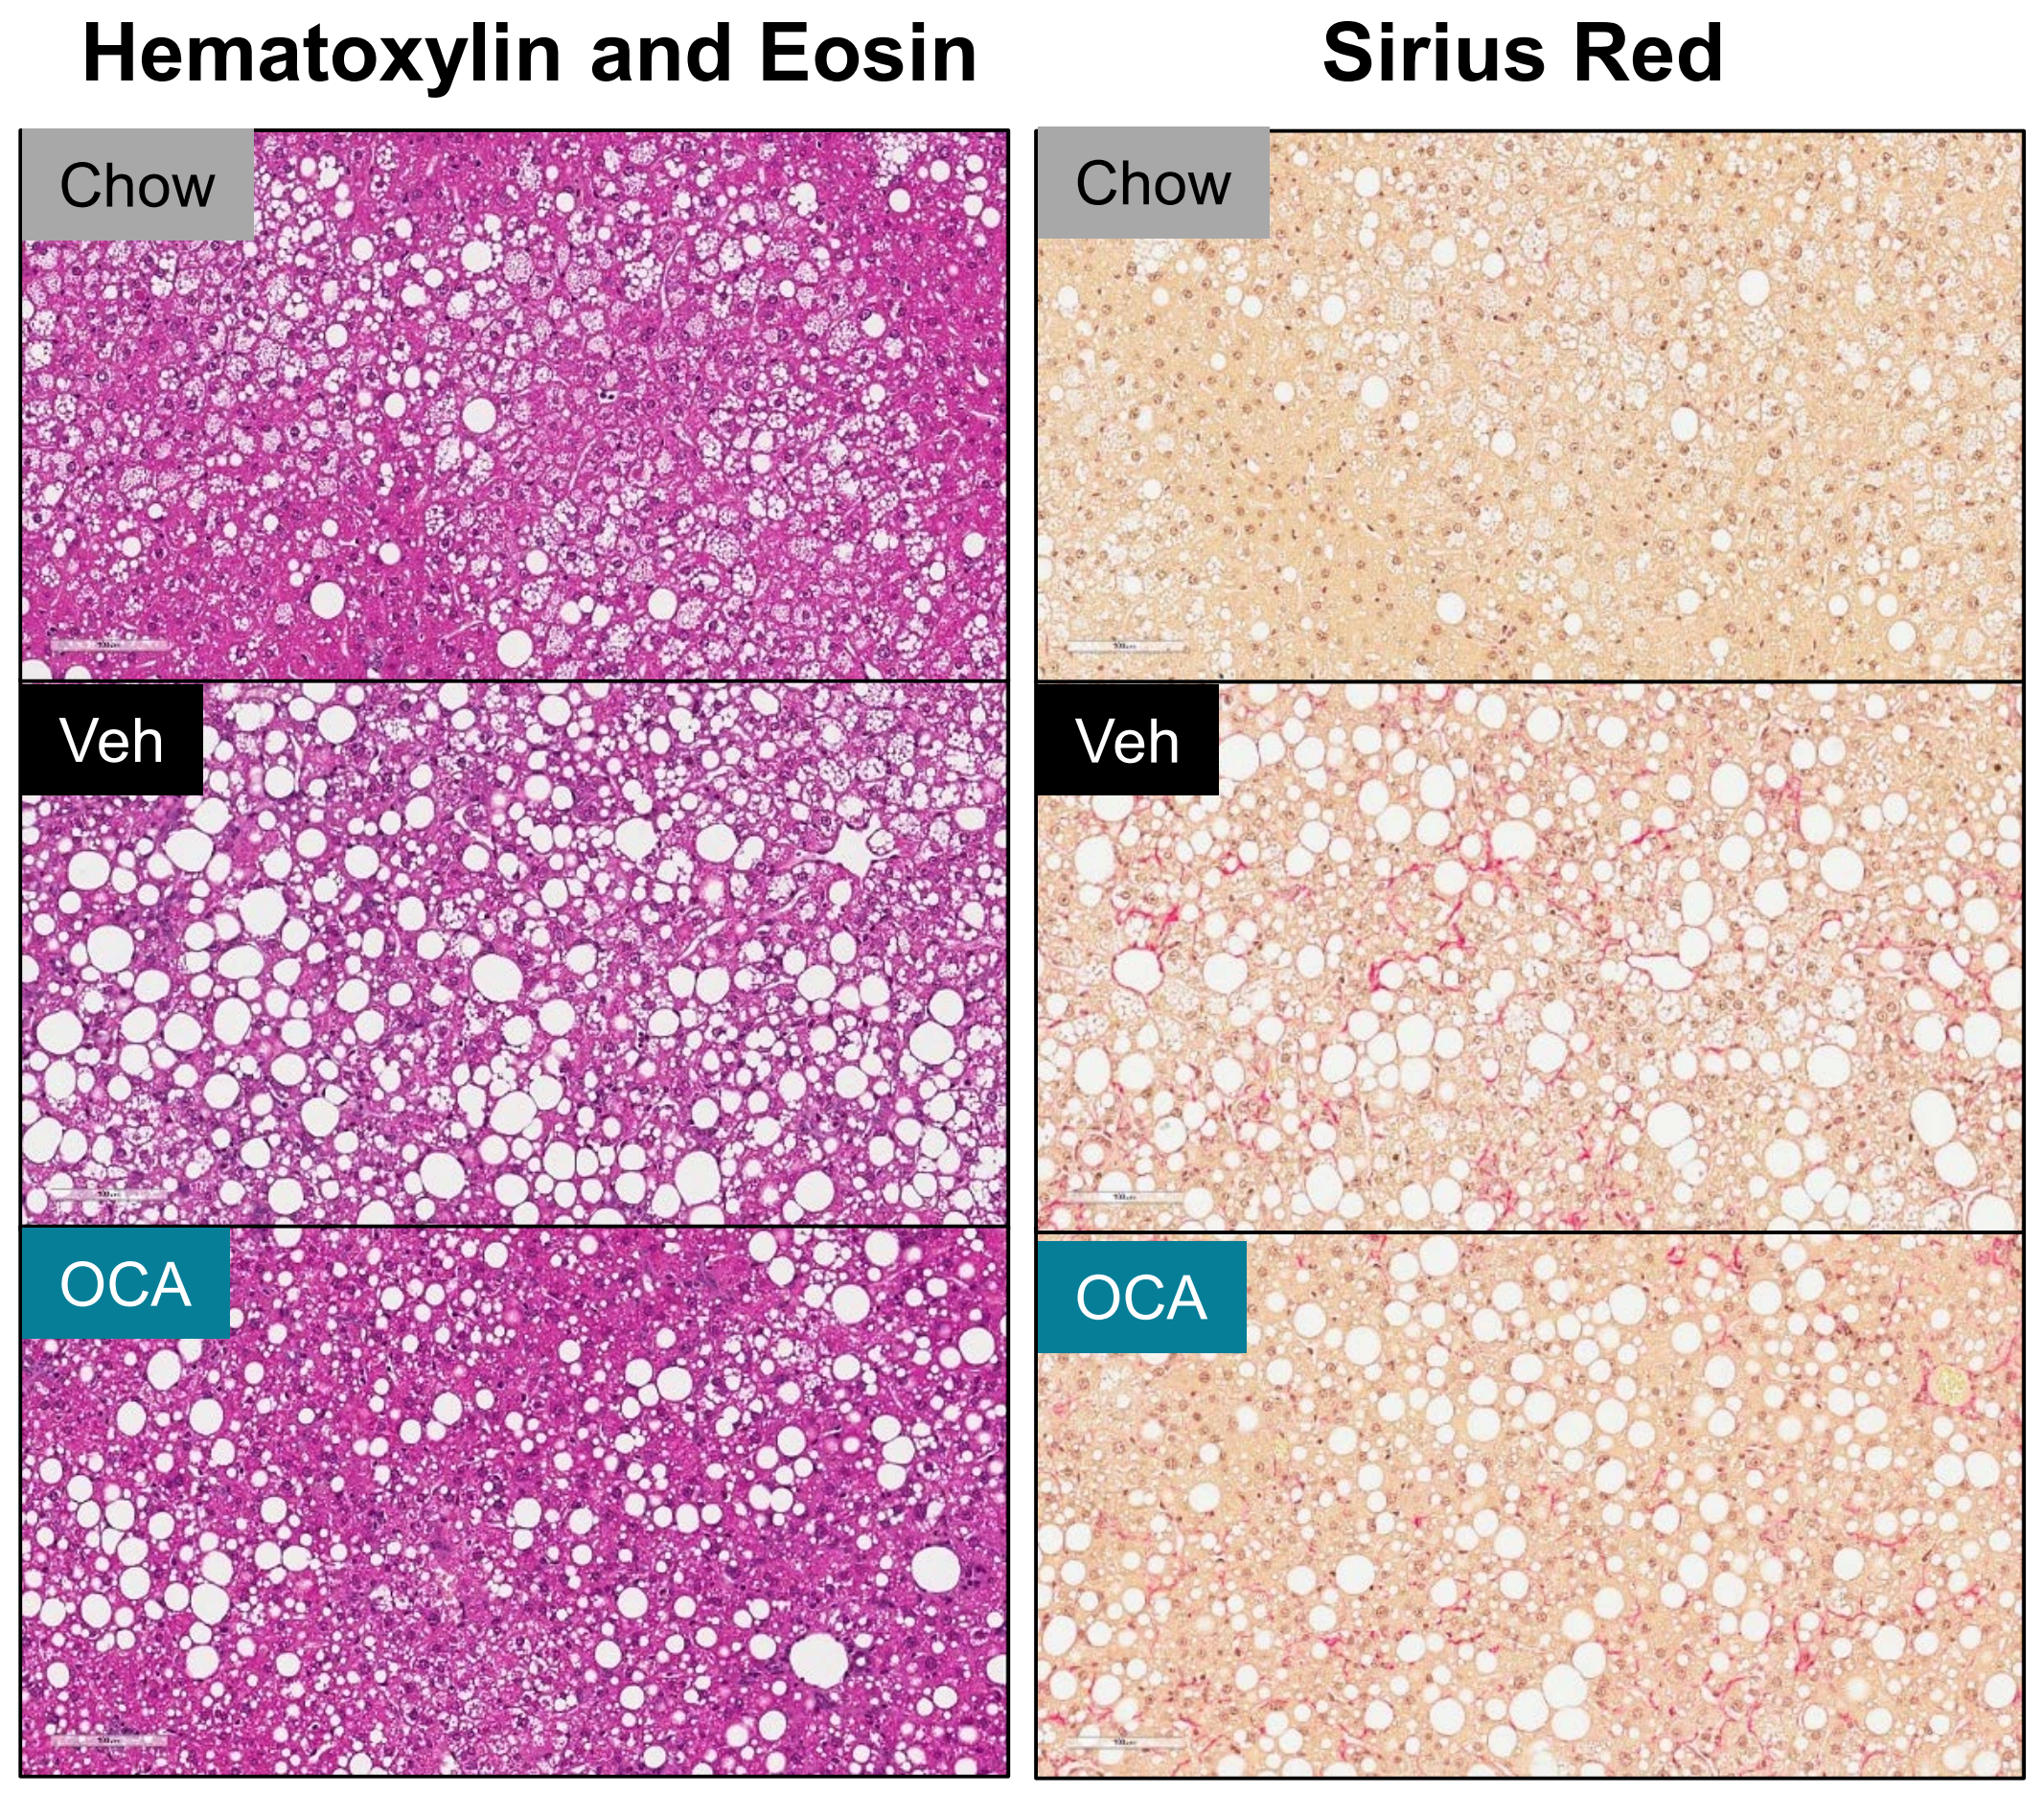

Hematoxylin and Eosin
Sirius Red
Chow
Veh
OCA

OCA Decreases NAFLD Activity Score†

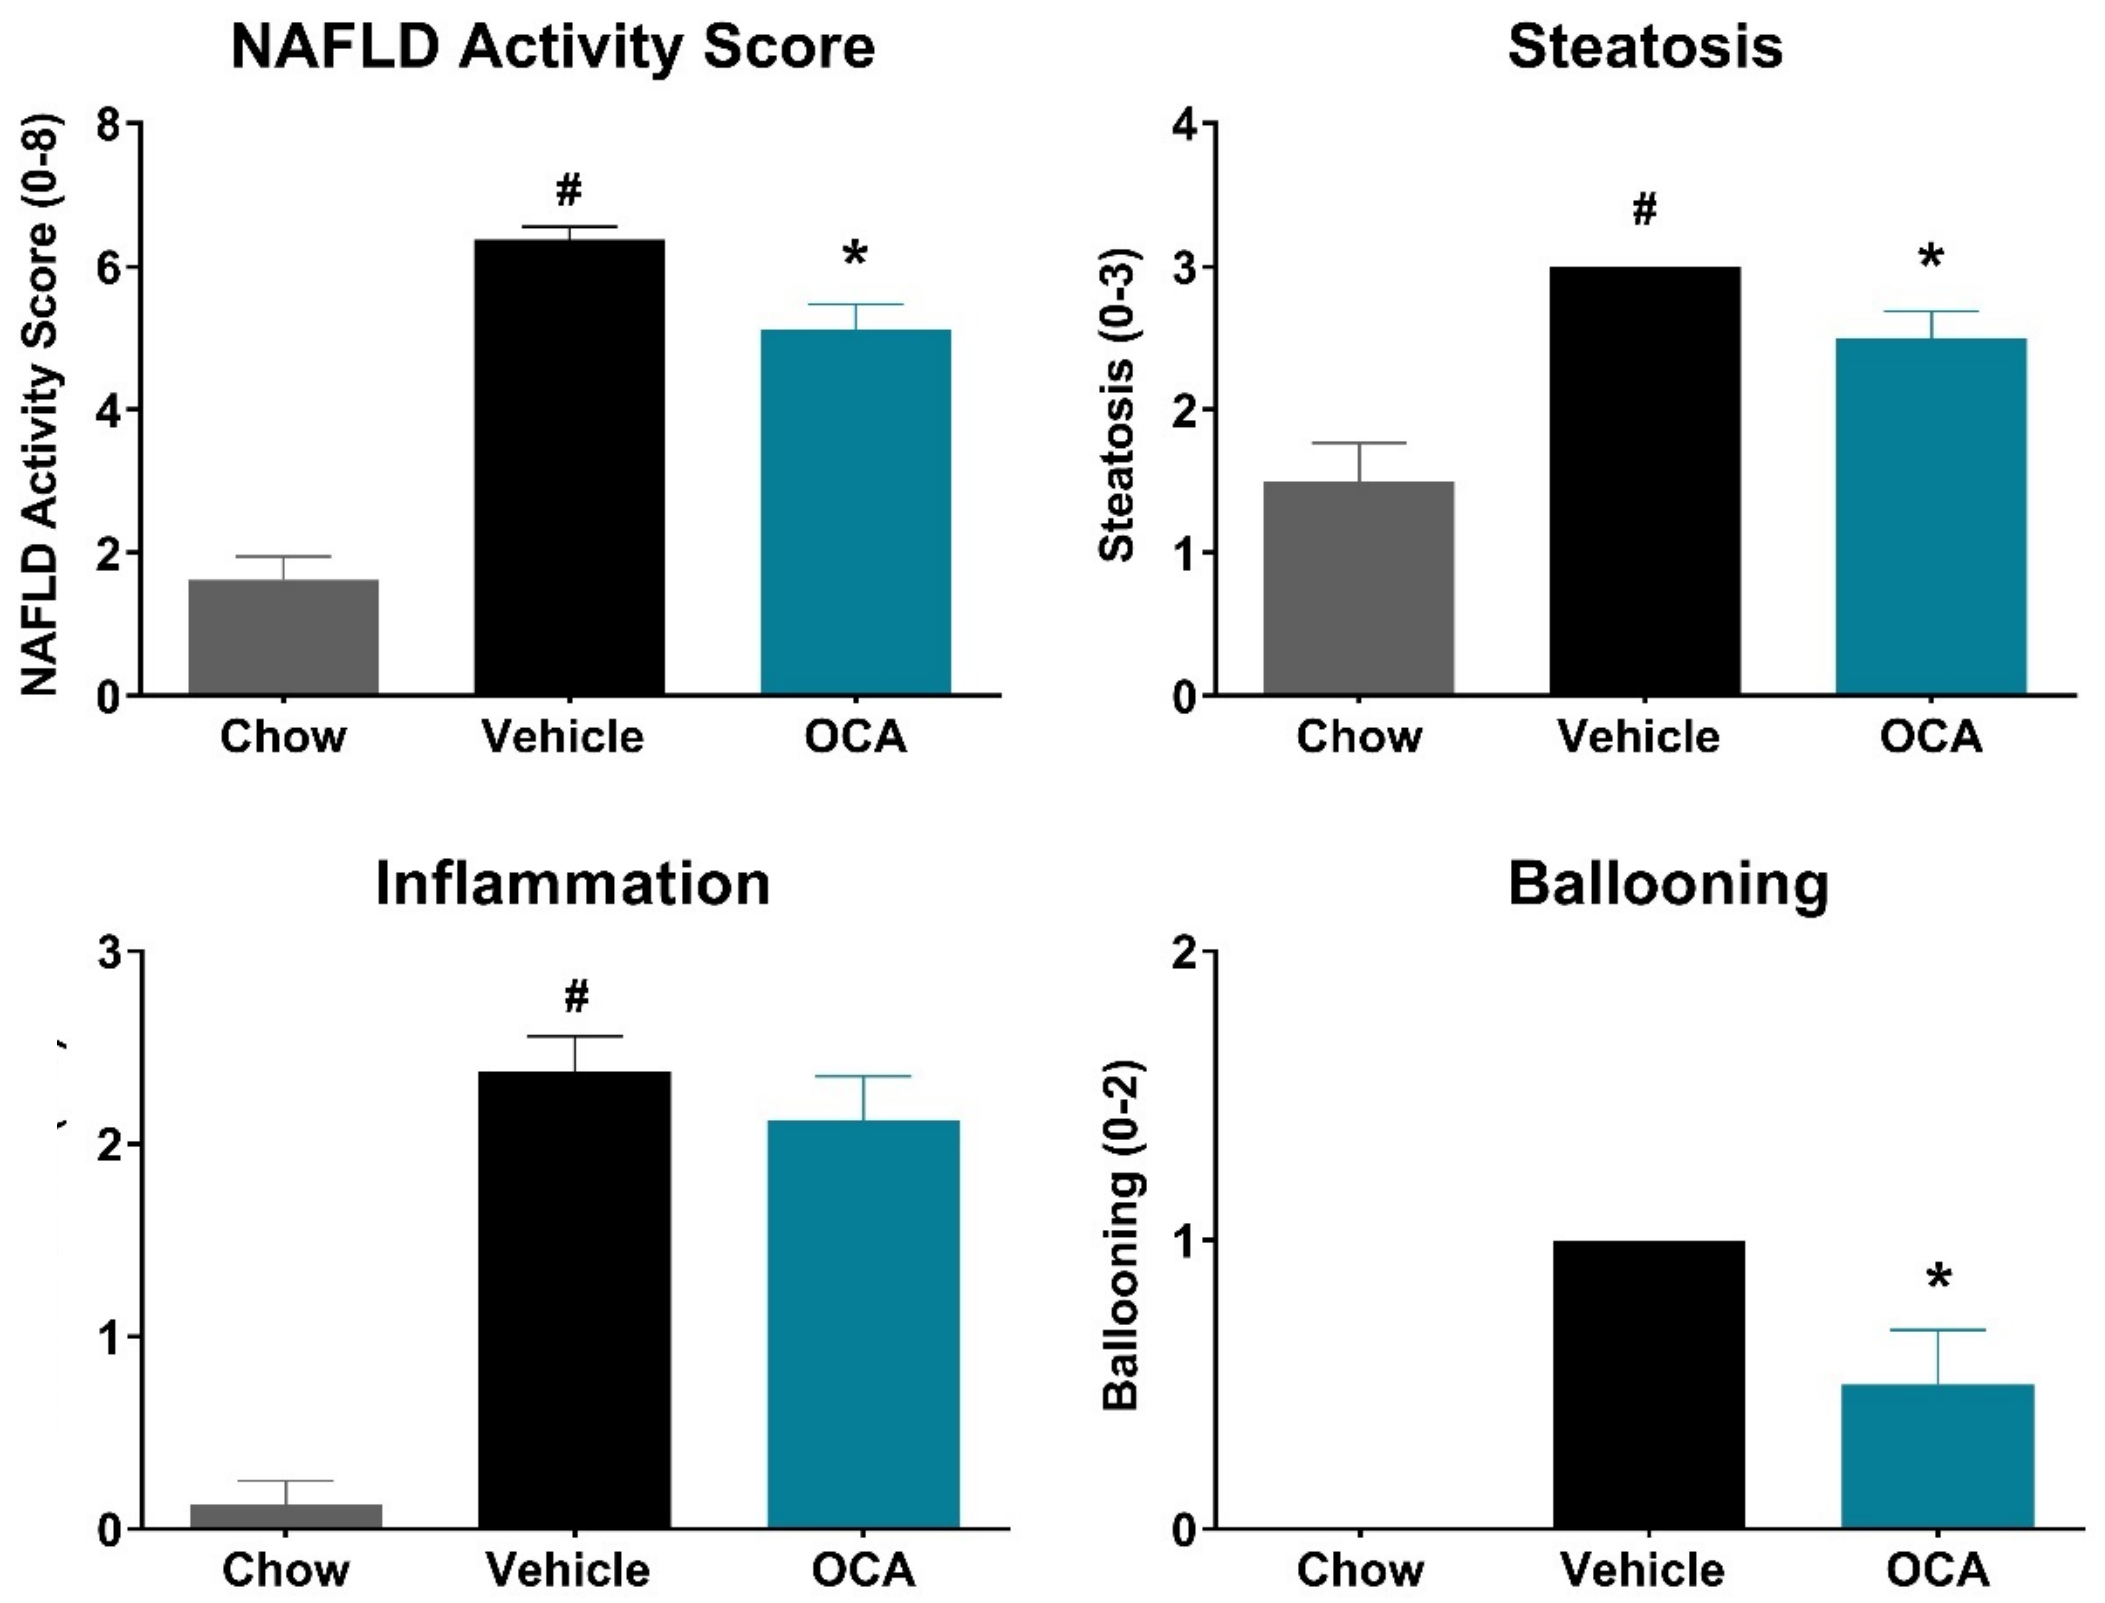

NAFLD Activity Score
NAFLD Activity Score (0-8)
Steatosis
Steatosis (0-3)
Inflammation
Ballooning
Ballooning (0-2)
Chow
Vehicle
OCA

OCA Decreases Steatosis†

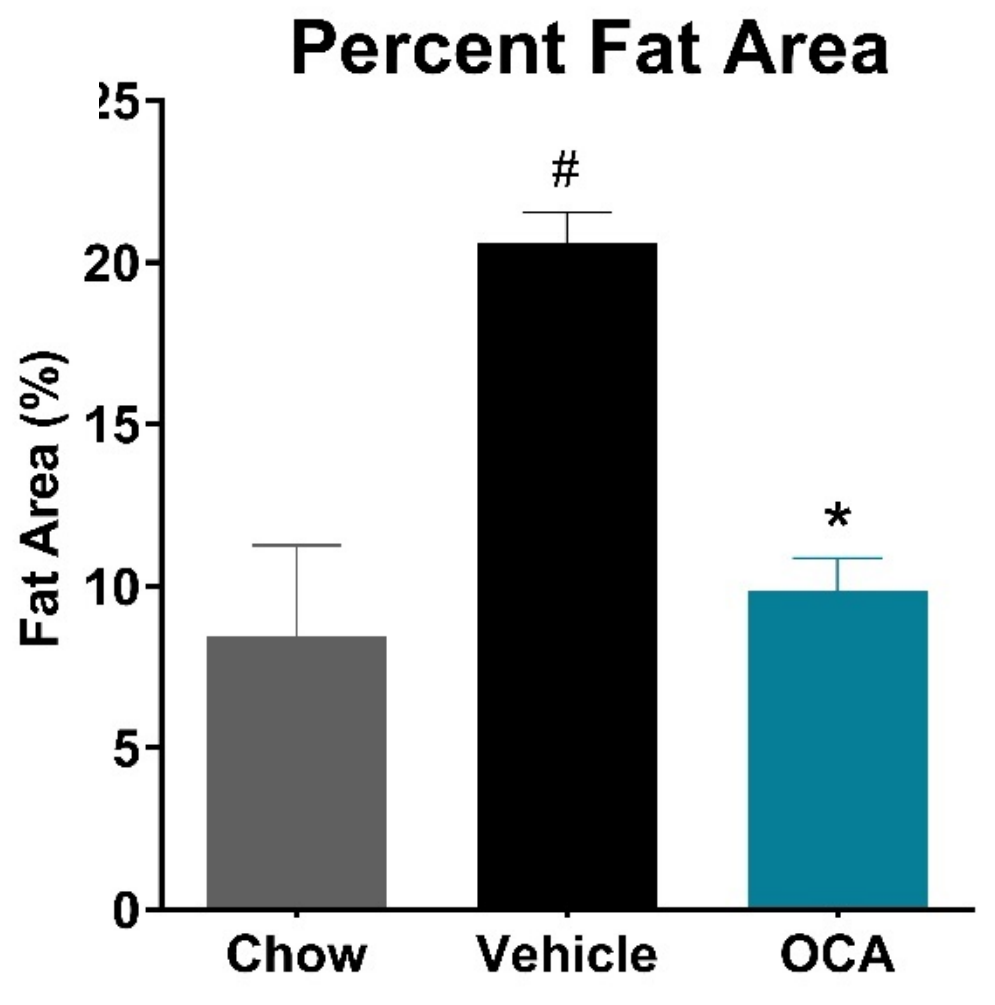

Percent Fat Area
Fat Area (%)
Chow
Vehicle
OCA

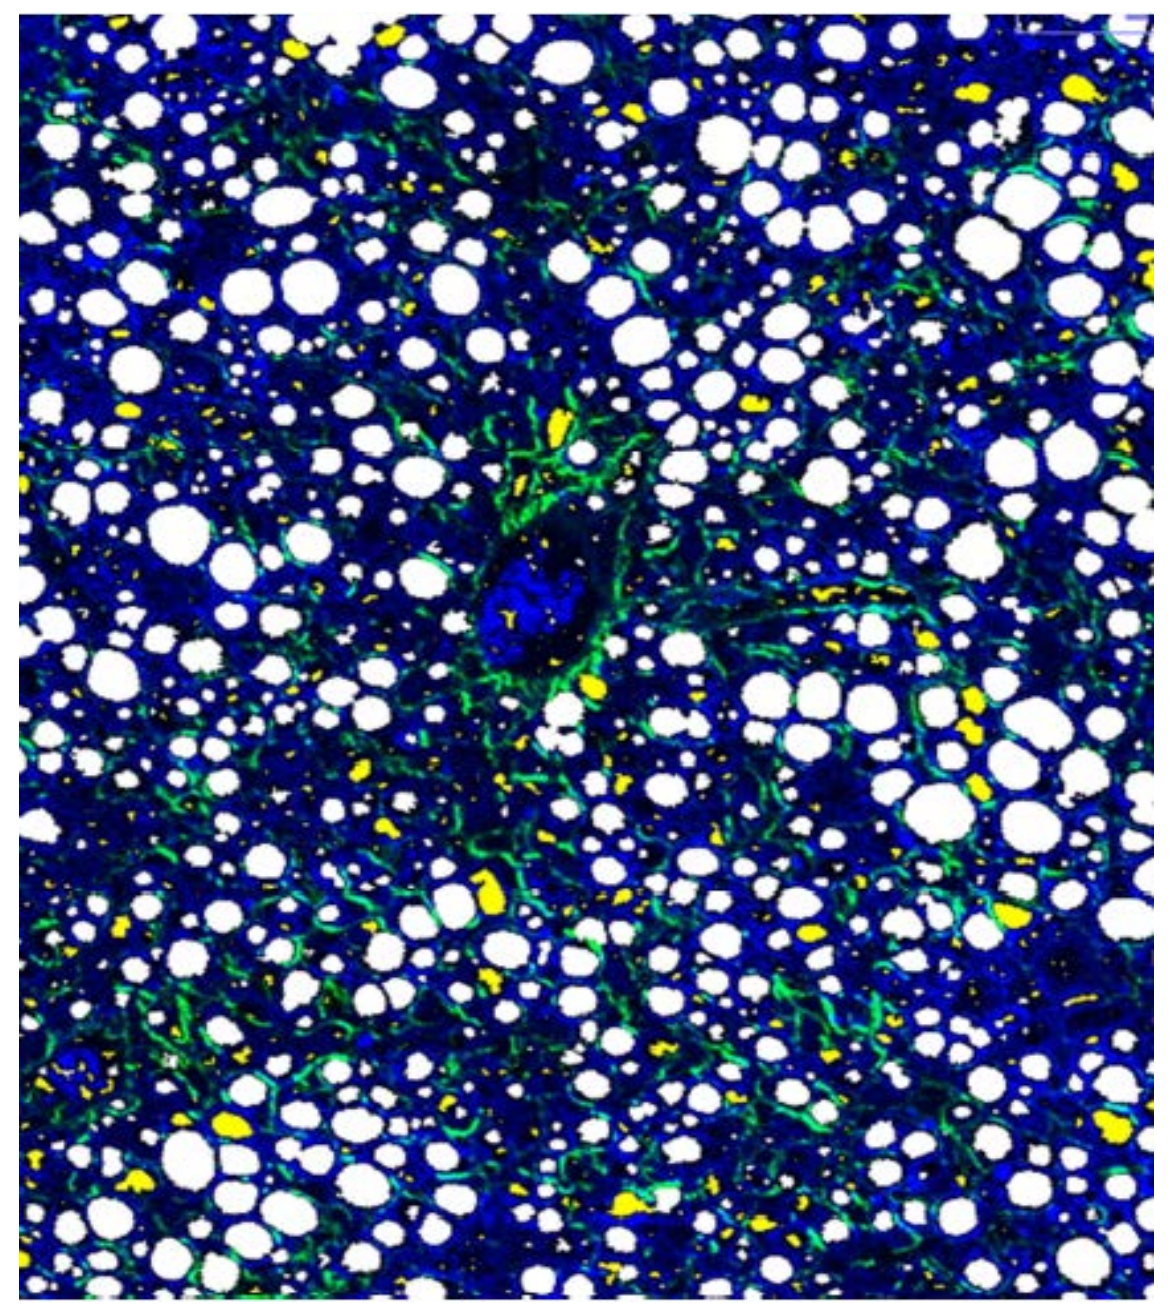

White = Fat Droplets
Yellow = Excluded
Green = Collagen
Blue = Auto-flourescence

Imaging performed by Genesis Imaging Services (Pharmanest).

†Data are Mean ± SEM; #p<0.05 Vehicle vs Chow; *p<0.05 OCA vs Vehicle; ANOVA with Dunnett's post-hoc.

OCA Decreases Fibrosis†

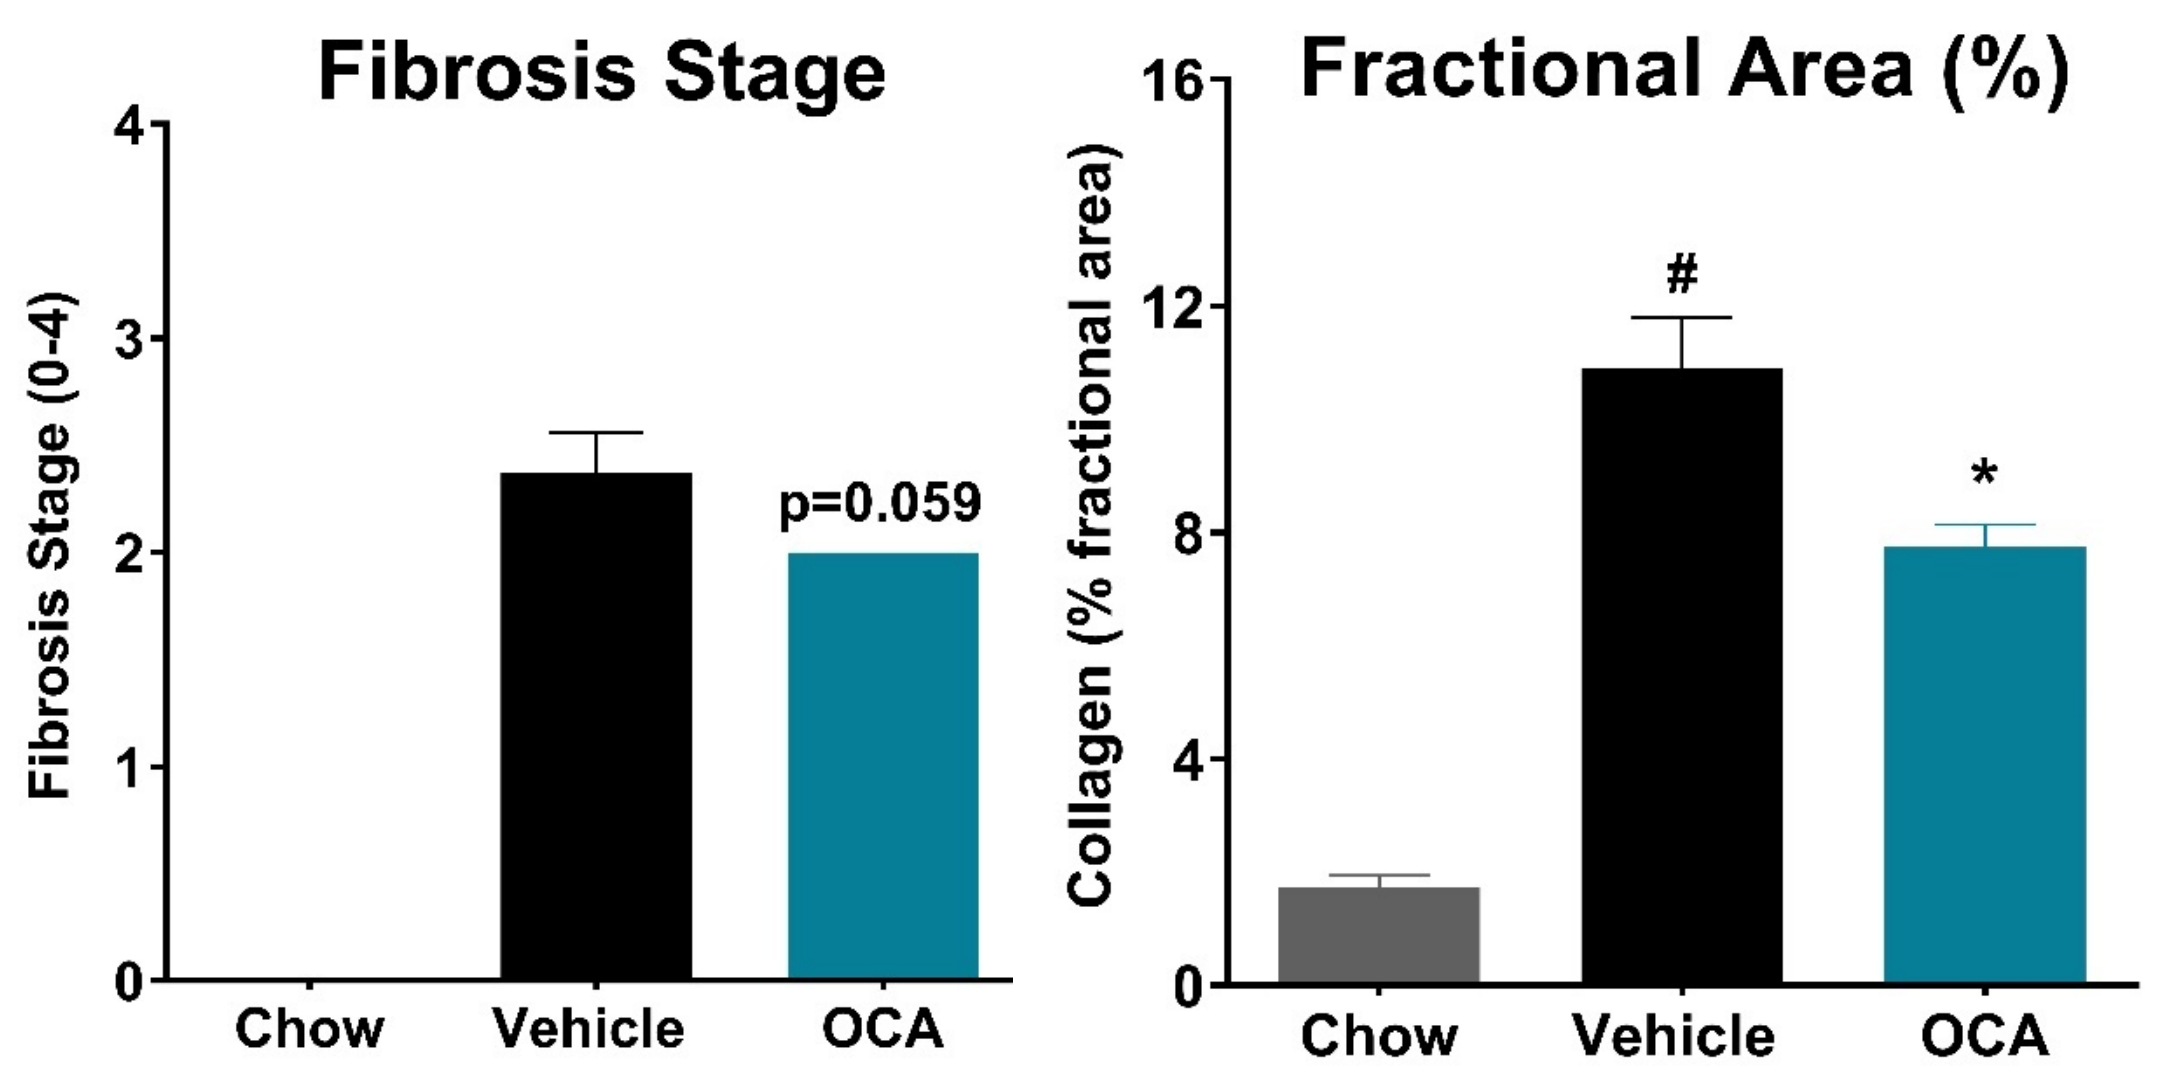

Fibrosis Stage
Fibrosis Stage (0-4)
p=0.059
Fractional Area (%)
Collagen (% fractional area)
Chow
Vehicle
OCA

OCA Decreases Collagen 1a1 (Fibrosis) by IHC†

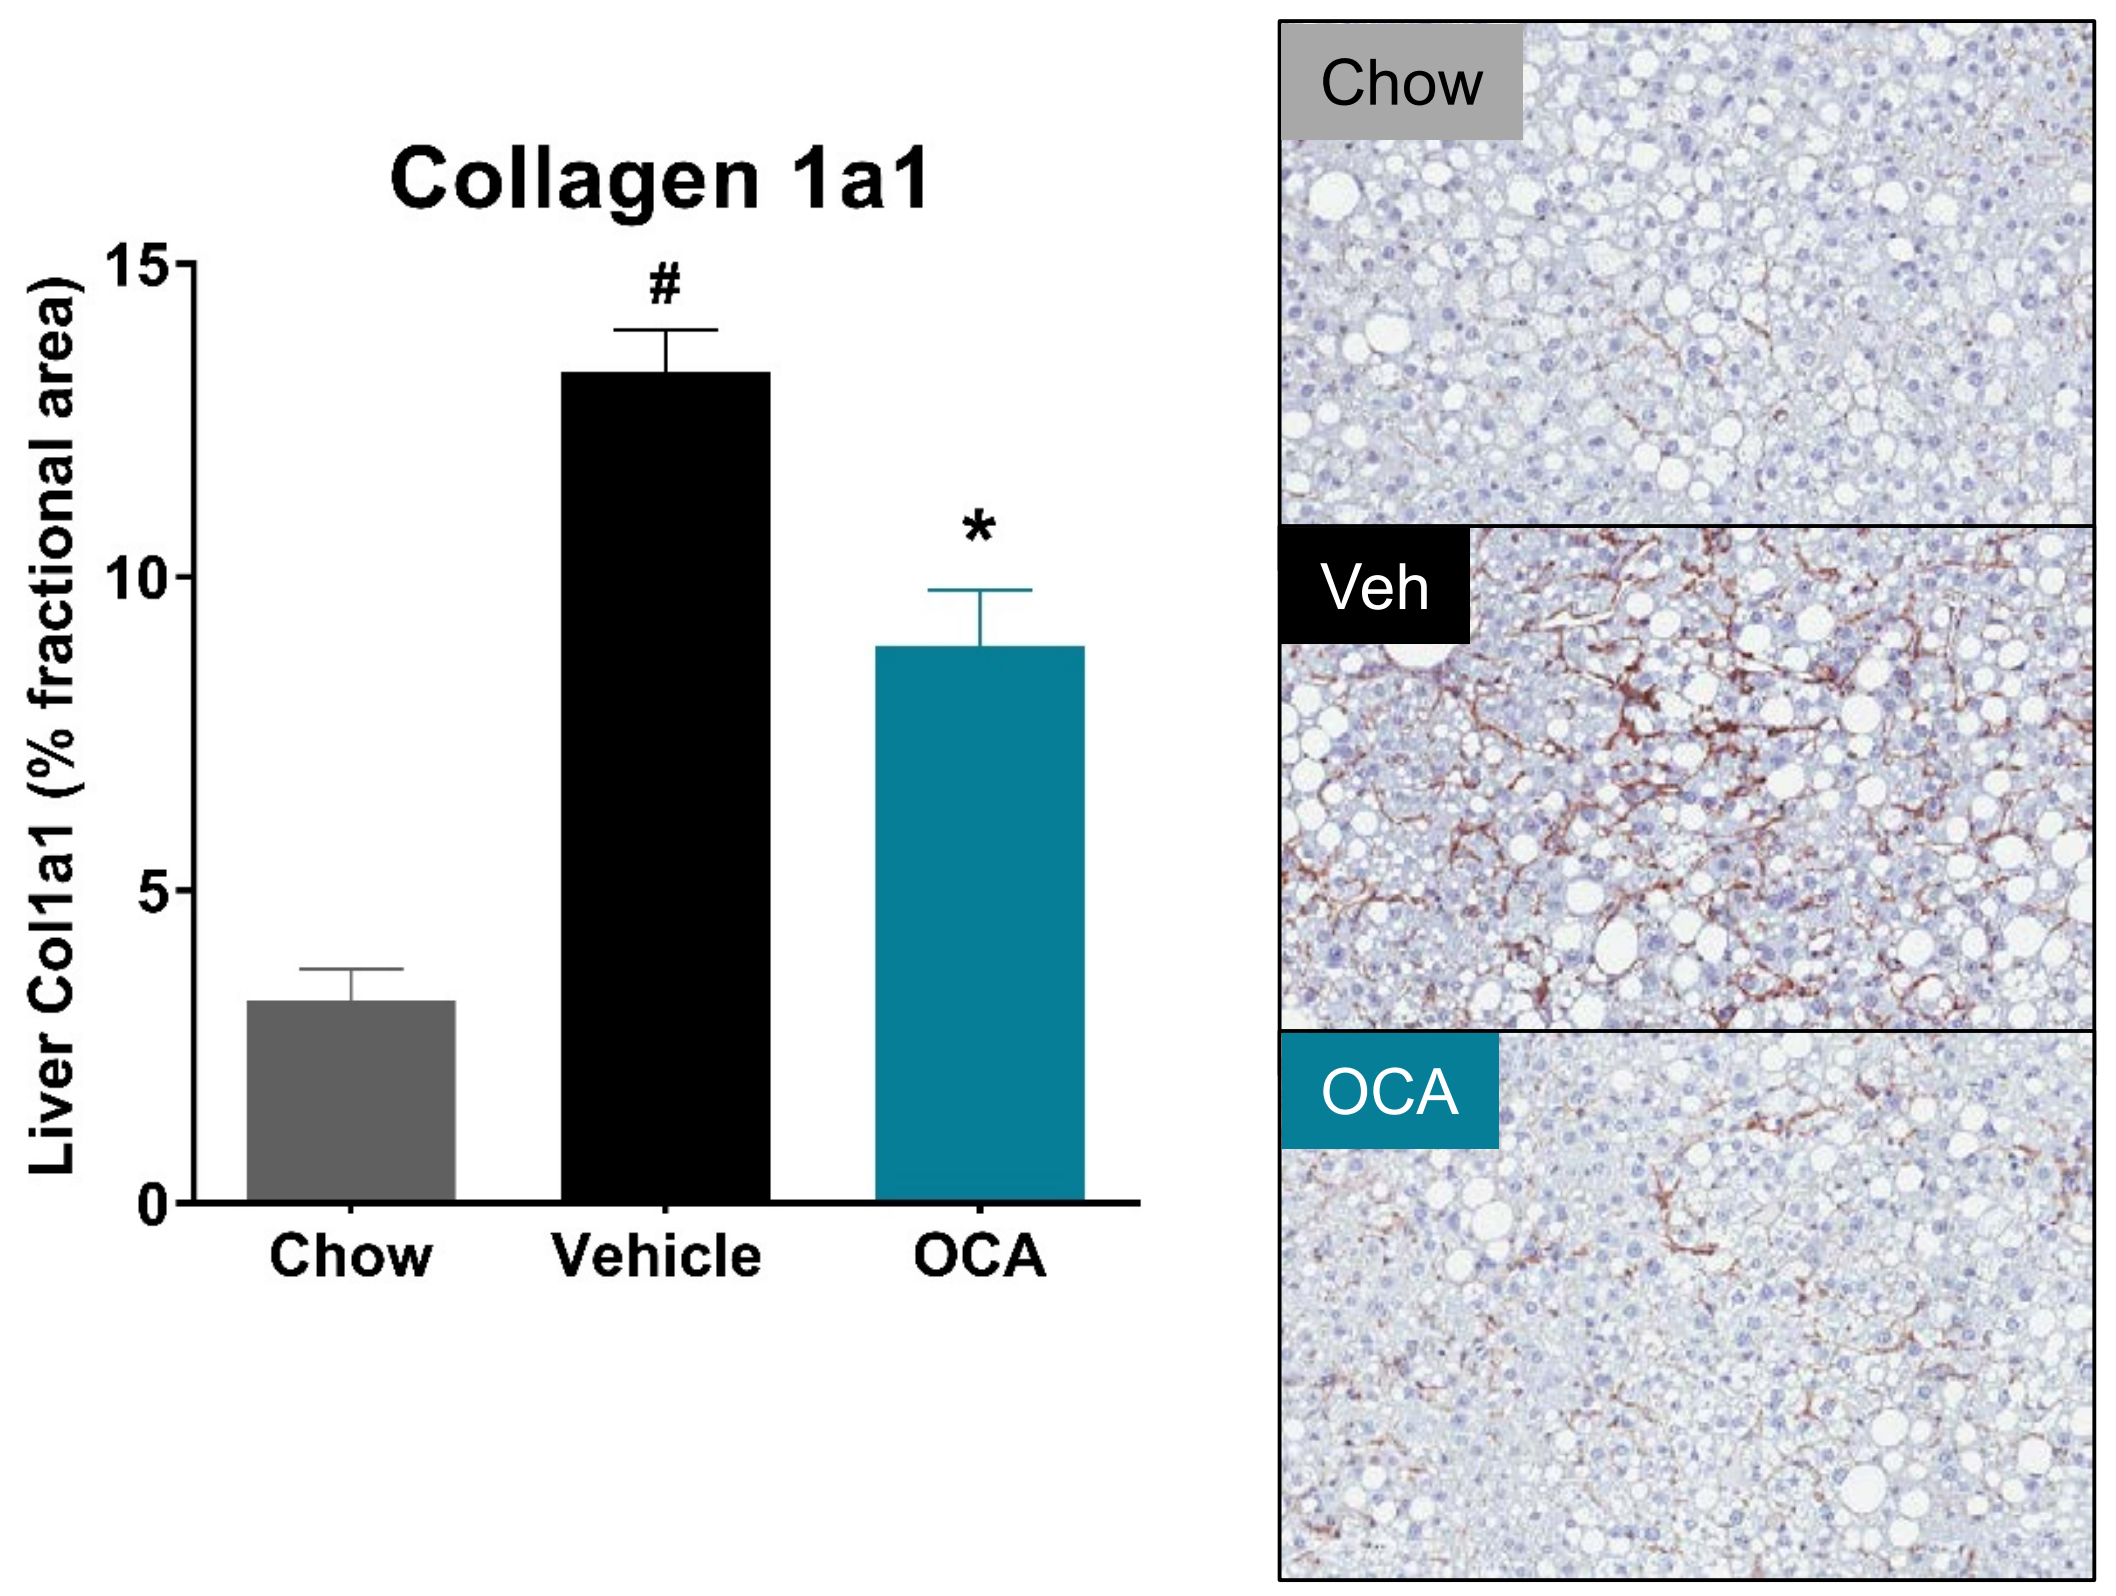

Collagen 1a1
Liver Col1a1 (% fractional area)
Chow
Vehicle
OCA
Chow
Veh
OCA

OCA Decreases Galectin-3 (Inflammation) by IHC†

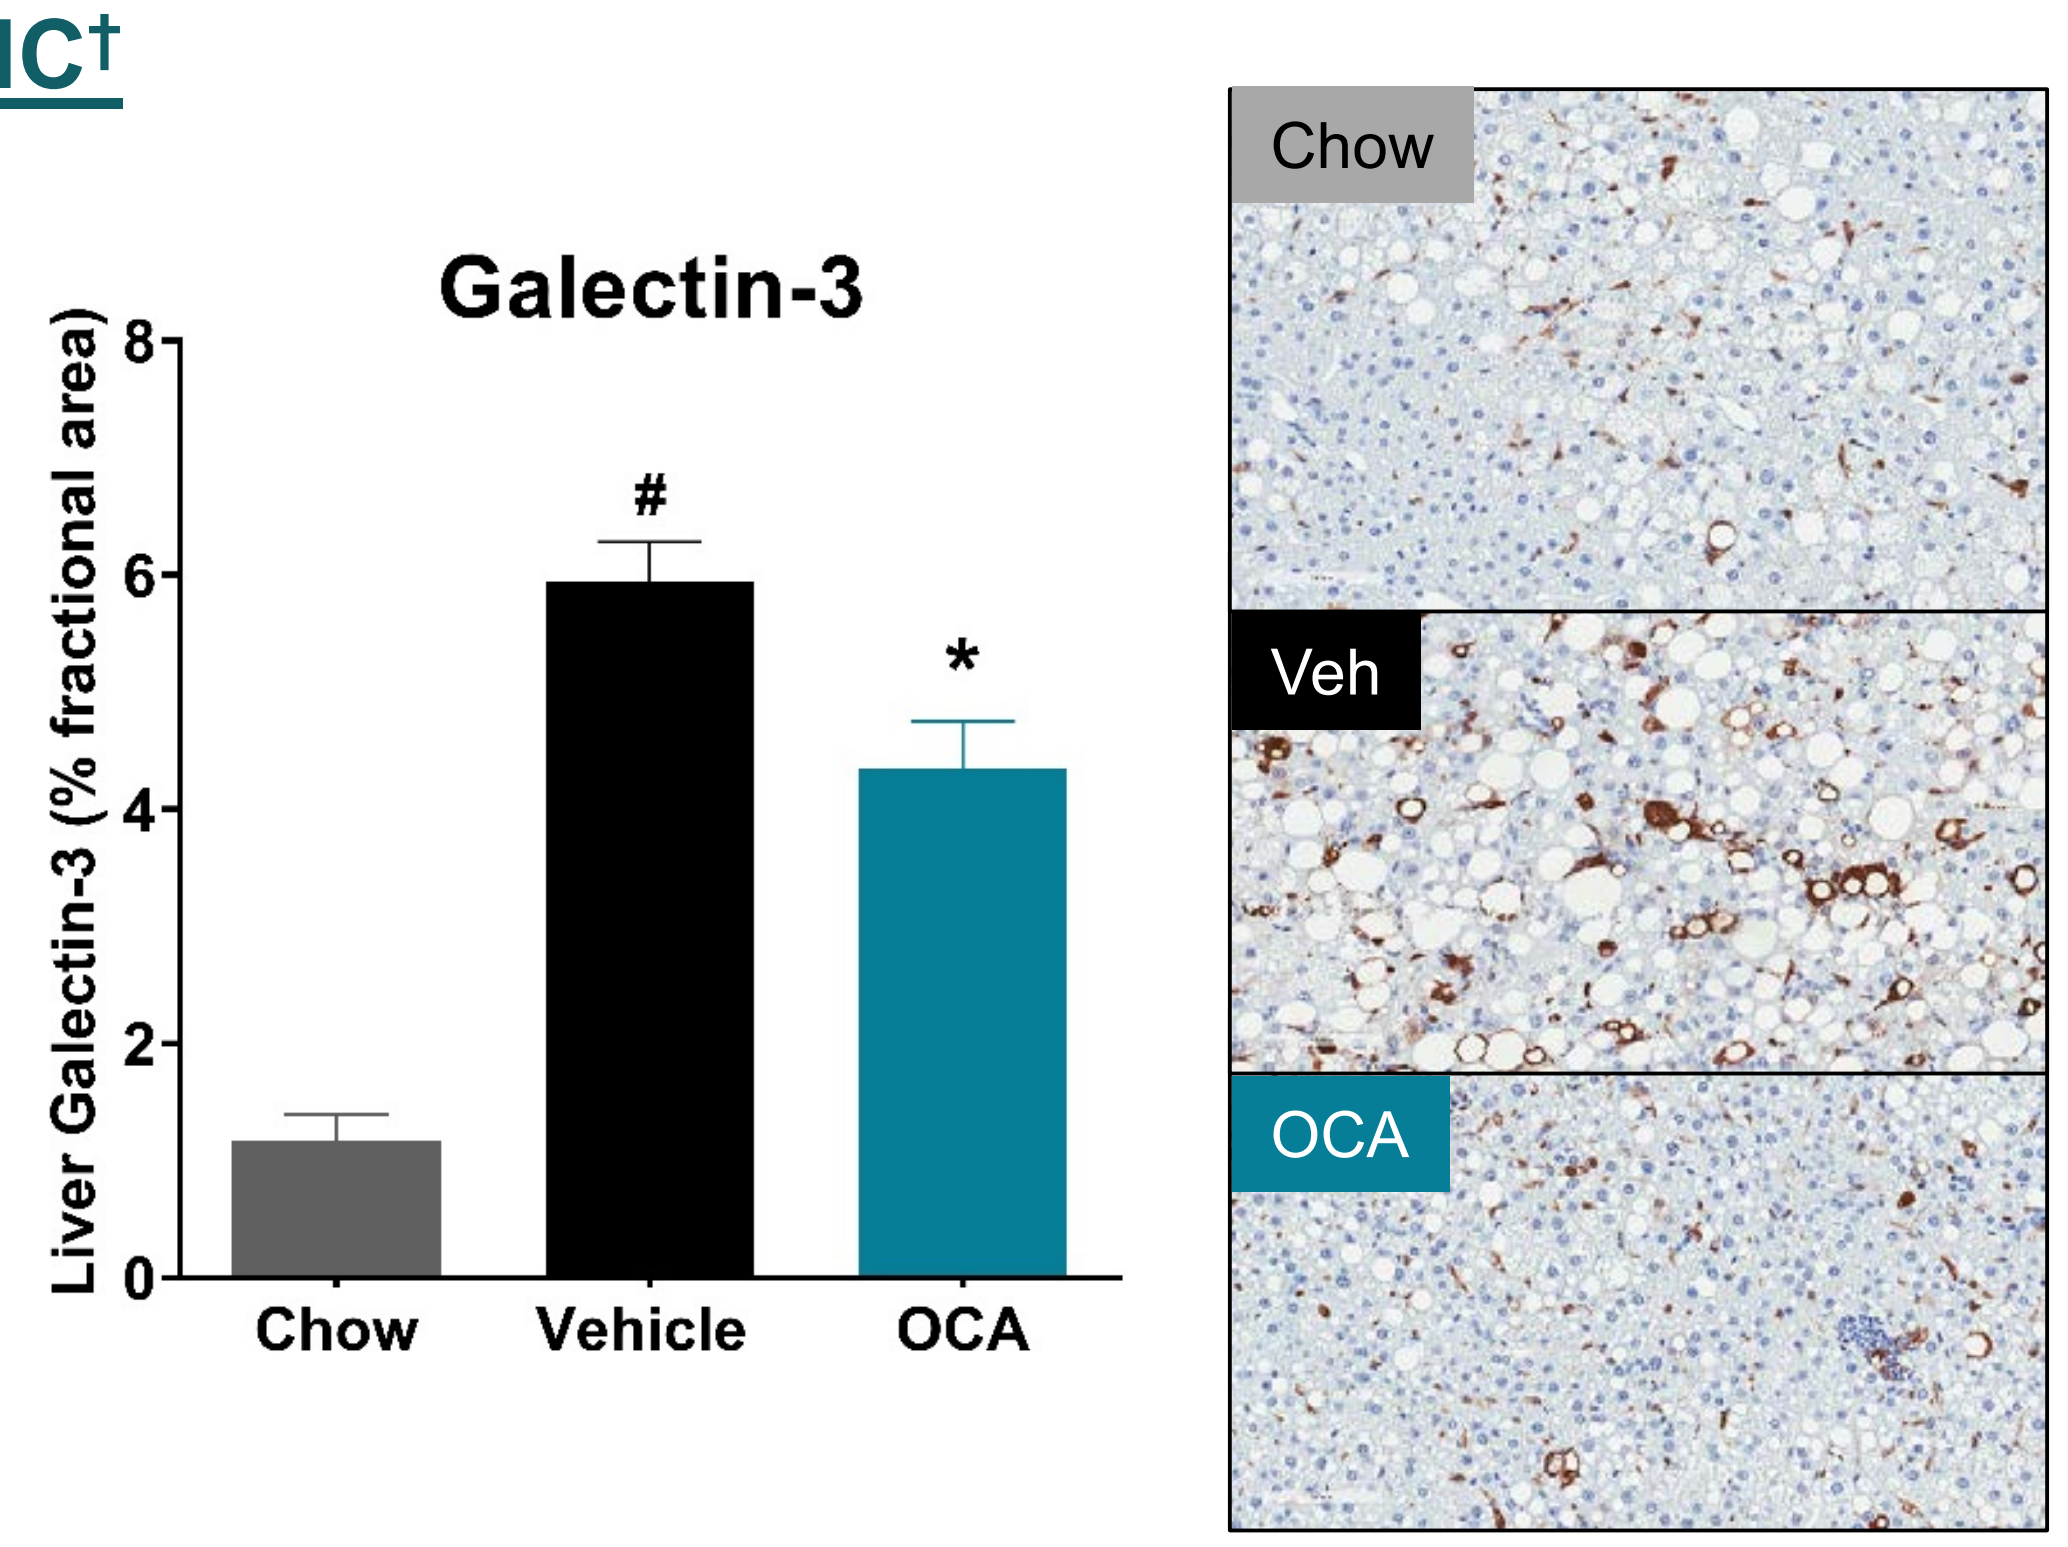

Galectin-3
Liver Galectin-3 (% fractional area)
Chow
Vehicle
OCA
Chow
Veh
OCA

†Data are Mean ± SEM; #p<0.05 Vehicle vs Chow; *p<0.05 OCA vs Vehicle, ANOVA with Dunnett's post-hoc.

OCA Alters Expression of FXR Signaling Genes in the Liver & Ileum†

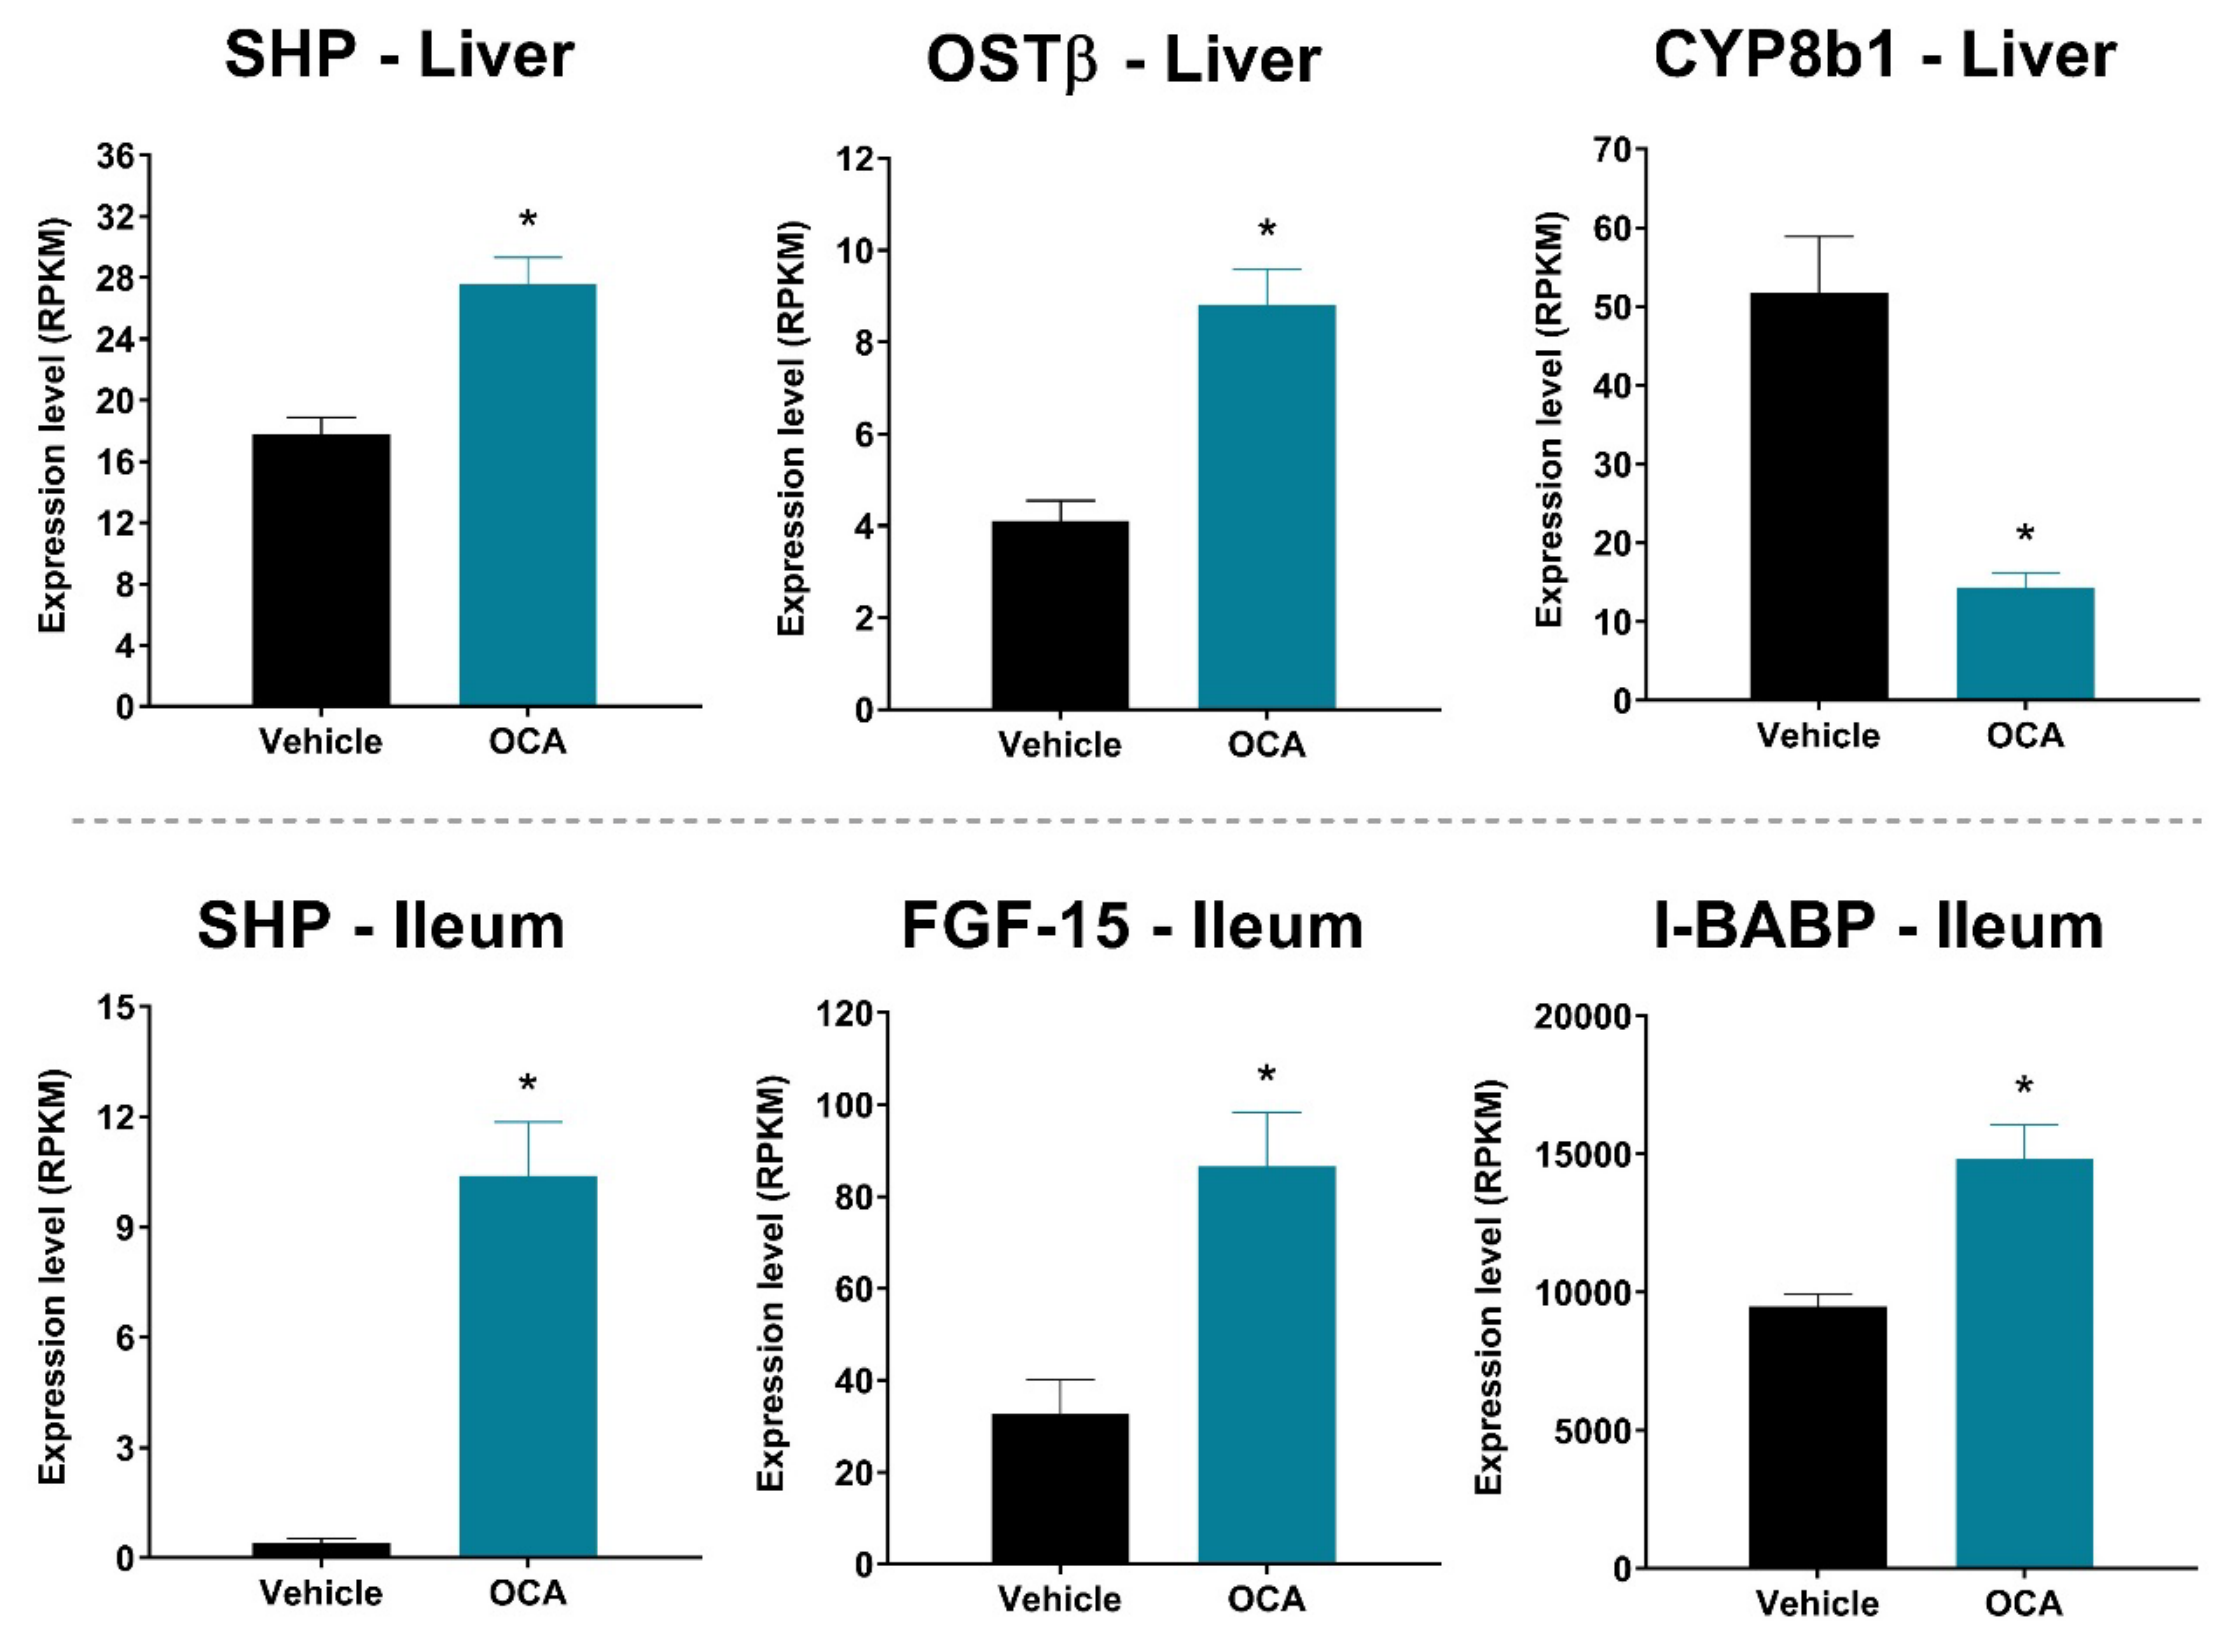

SHP - Liver
OSTβ - Liver
CYP8b1 - Liver
SHP - Ileum
FGF-15 - Ileum
I-BABP - Ileum
Expression level (RPKM)
Vehicle
OCA

Effects of OCA on Disease-related Genes in the Liver†

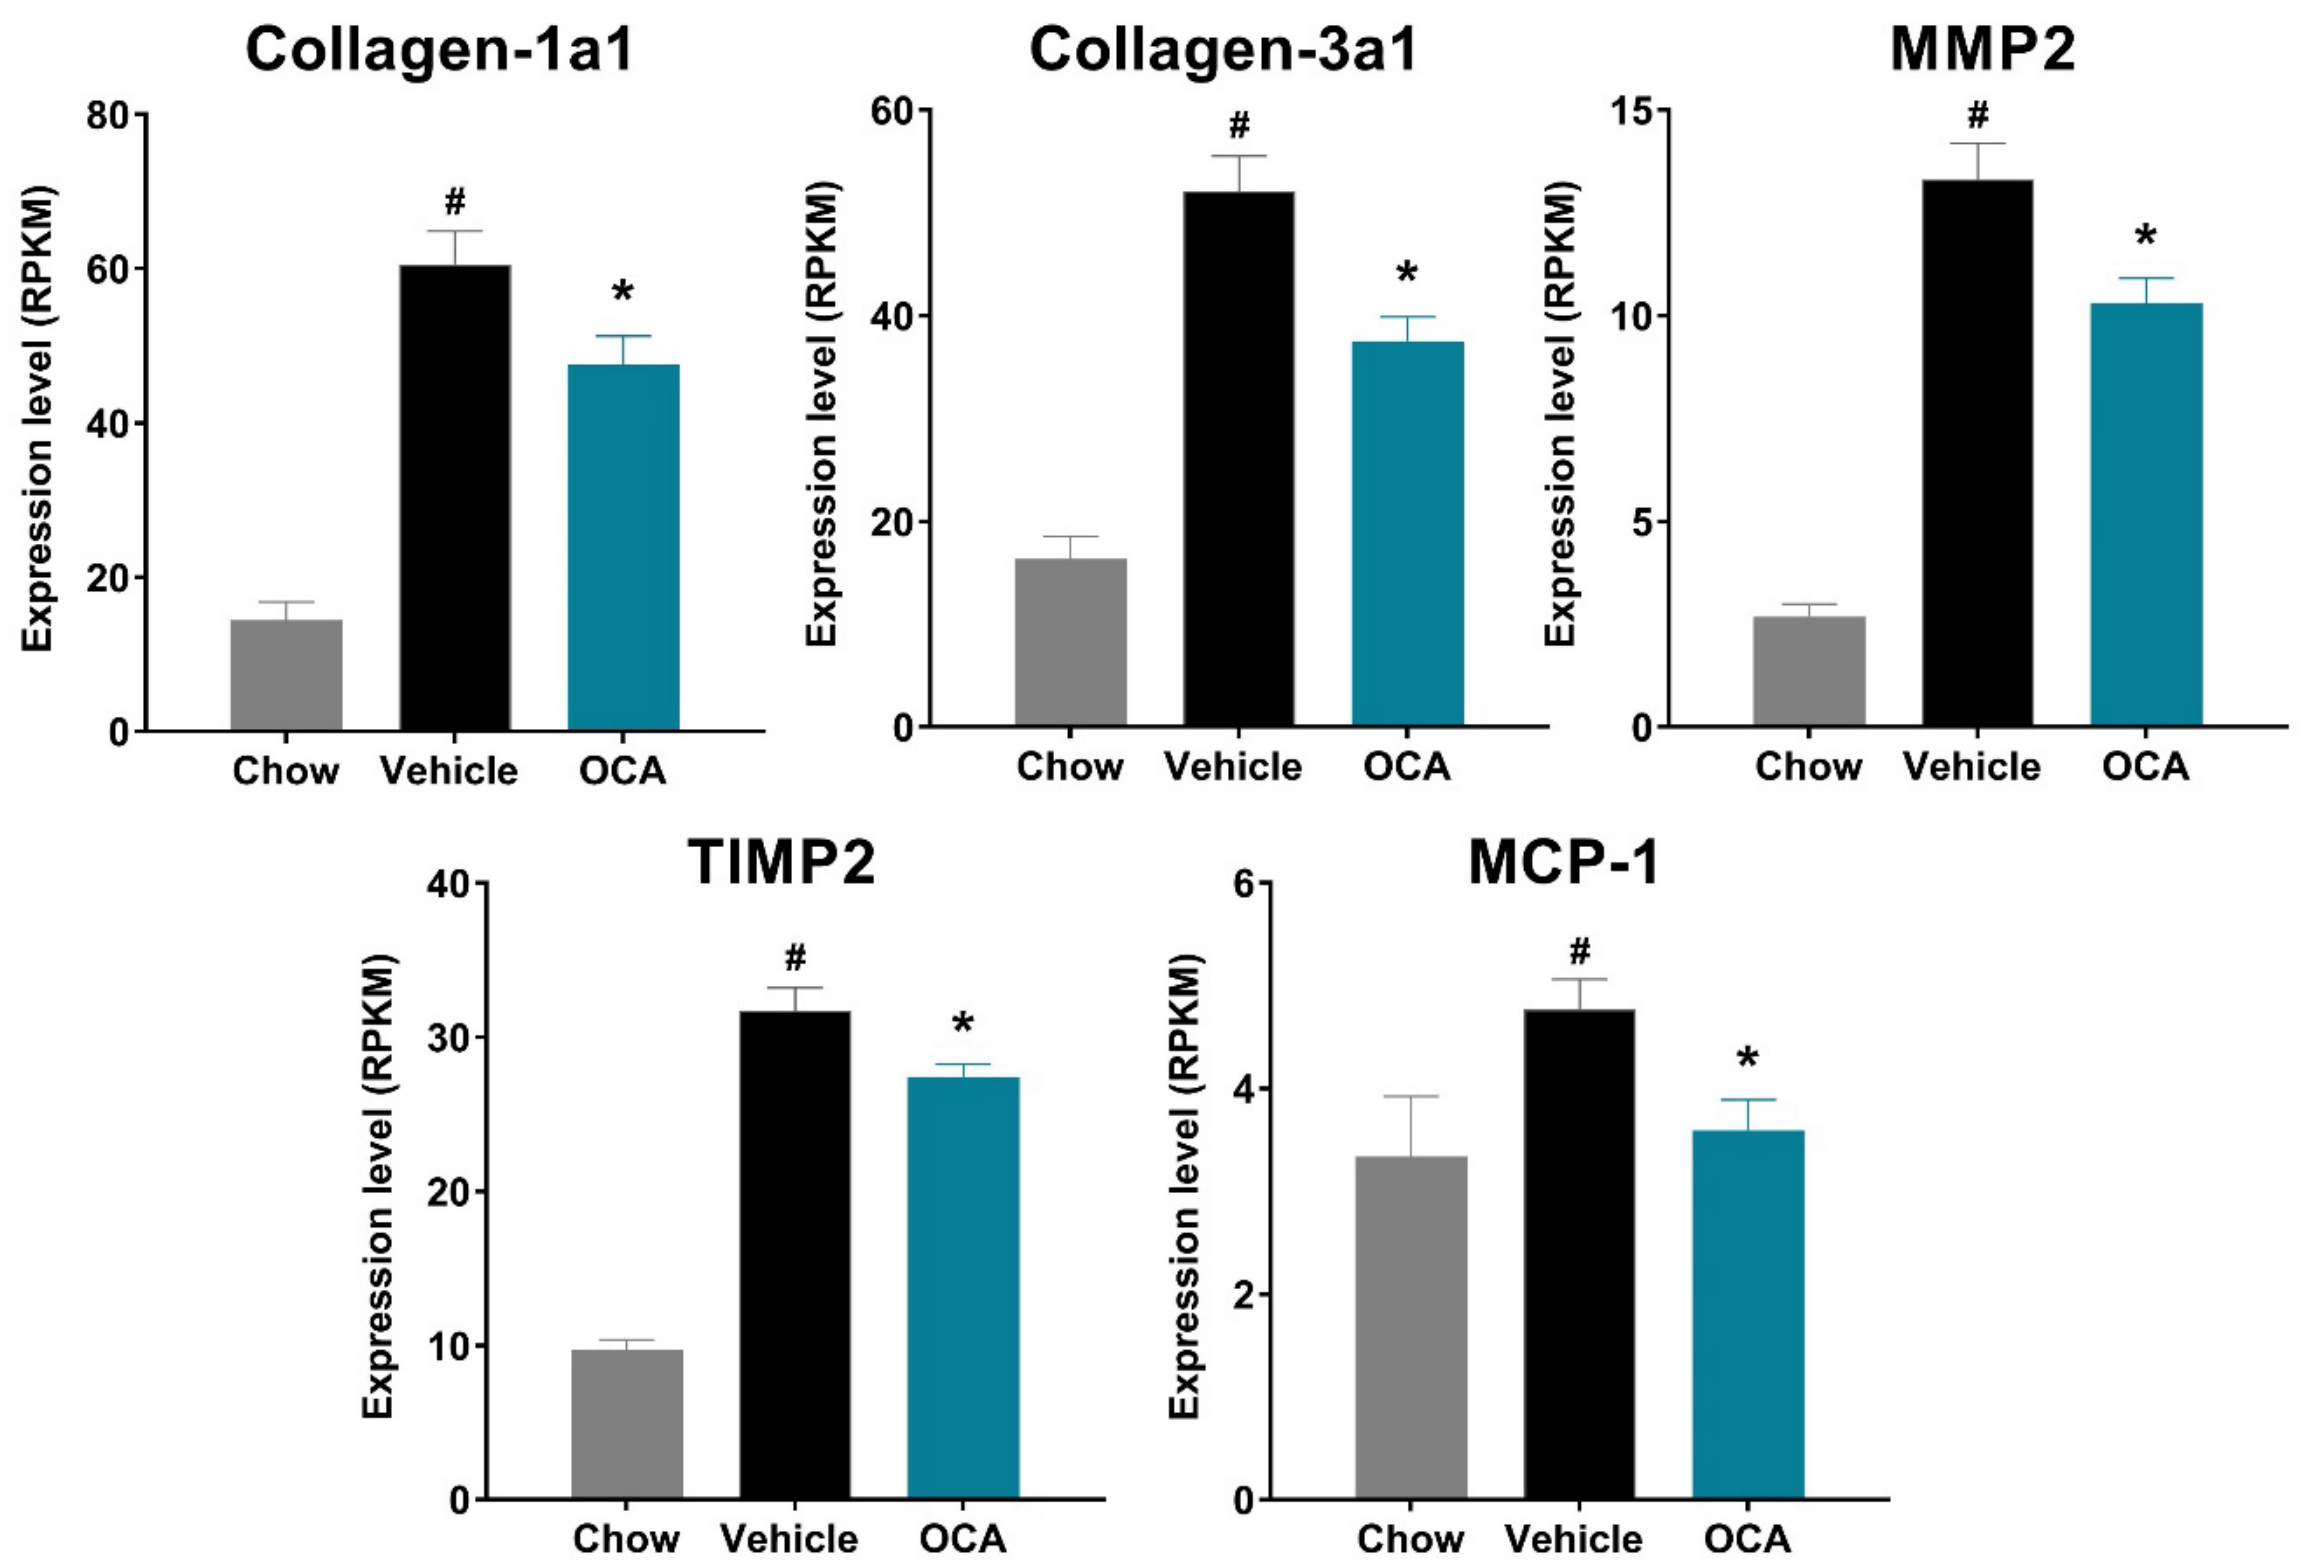

Collagen-1a1
Collagen-3a1
MMP2
TIMP2
MCP-1
Expression level (RPKM)
Chow
Vehicle
OCA

†Data are Mean ± SEM; #p<0.05 Vehicle vs Chow; *p<0.05 OCA vs Vehicle, ANOVA with Dunnett's post-hoc.

DISCLAIMER

OCA is indicated in the US and EU for the treatment of PBC in combination with UDCA in adults with an inadequate response to UDCA, or as monotherapy in adults unable to tolerate UDCA. While OCA has received marketing authorization from the FDA and the European Commission for the treatment of PBC, this drug has not been approved for use by any other regulatory bodies at this time. No conclusions can be drawn concerning the safety or efficacy of OCA in indications other than PBC or in areas outside the US and EU.

CONCLUSIONS

- Obeticholic acid improved histological indices of NASH as assessed by NAS scoring and immunohistochemical methods in Gubra AMLN mice
- Improvements were associated with activation of key FXR signaling genes in the liver and ileum and salutary effects on markers of inflammatory and fibrotic processes
- These findings further confirm the benefits of FXR agonism in a rodent model of diet-induced and biopsy-confirmed NASH

ACKNOWLEDGEMENTS

Mathieu Petitjean, PhD and Li Chen, PhD – Genesis Imaging Services (Pharmanest)

Presented at EASL, The International Liver Congress, April 19-23, 2017, Amsterdam, The Netherlands